Approaches to Detect Non-Compliant Patients and Obtain Unbiased Estimates of Population PK Model Parameters in a Population with Prevalent Non-Compliance

Leonid Gibiansky[1], Ekaterina Gibiansky[1], Valerie Cosson[2], Nicolas Frey[2], Franziska Schaedeli Stark[2]

[1]QuantPharm LLC, North Potomac MD, USA; [2]F. Hoffmann-La Roche Ltd, Basel, Switzerland

ACoP 2013

BACKGROUND

Non-compliance is a significant issue for many drugs with chronic outpatient administration.

- Most estimation methods are sensitive to outliers. Even small fraction of non-compliant patients may significantly affect population pharmacokinetic (PK) parameter estimates.
- There are no commonly accepted and tested methods to identify non-compliant patients and obtain unbiased estimates of population PK parameters.

OBJECTIVES

To propose and evaluate methods for detection of non-compliance using concentration-time data and for obtaining unbiased estimates of population PK model parameters in a population with prevalent non-compliance.

METHODS

Method CM1:

Ref. [1] proposed to detect subjects with odd observations and reduce their influence on the population PK parameter estimates by introduction of the random effect on residual error.

We apply the same idea to detection of non-compliant patients that can be distinguished by strong fluctuations of observed concentration levels that could not be explained by the model that assumes full compliance and time-independent parameters.

<u>The proposed procedure</u>:

- Fit the model with the random effect η_ε on the residual error;
- Identify subjects with strong non-compliance as those with high η_ε;
- Exclude non-compliant subjects from the dataset to obtain unbiased estimates of model parameters.

Method CM2:

Ref. [2] proposed to account for non-compliance using only data that follow the in-patient (compliant) doses:

- Restricted to a one-compartment model with absorption half-life much shorter than inter-dose interval
- In this case, concentrations that follow in-patient doses are $C(t)=C_0\exp(-k_e t)+A[\exp(-k_e t)-\exp(-k_a t)]$, and the parameters ($C_0$, A, k_e, k_a) can be estimated from only the reliable data that follow the in-patient dose.

We propose:

- Extension of this method to the multi-compartment models with absorption and distribution half-life much shorter than inter-dose interval;
- Simpler implementation that can be applied to both linear models with known analytical solution and to non-linear models.

<u>The proposed procedure</u>:

- Use only data of samples preceding and following the in-patient doses
- Introduce individual relative bioavailability (with high and fixed variance) for outpatient doses. If more than one sampling period with in-patient doses is available, allow separate bioavailability parameters for outpatient doses preceding each of these periods.

Advantages and limitations:

- Can be applied to both linear and non-linear models with absorption and distribution half-life much shorter than inter-dose interval
- Simple to implement without analytical solution of underlying equations
- Rely on the availability of sufficient data following in-patient doses
- For one-compartment linear model, reduces to the method proposed in [2]

SIMULATED DATA

<u>Model</u>: two-compartment linear; QD administration; relatively long half-life and significant drug accumulation.

<u>Non-compliance patterns</u>:

- 30% - 50% or 100% non-compliers;
- 10 - 80% of randomly chosen doses associated with 1-6 days drug holidays.

<u>Subjects and samples</u>:

- CM1 method: sparse data from 400 subjects;
- CM2 method: rich data from 200 subjects;
- CM1 method and combined CM1/CM2 methods: combination of 80%/20% subjects with sparse/rich data, respectively.

RESULTS

- For the models that did not account for non-compliance, parameter estimates were biased, with bias in clearance approximately equal to the fraction of missed doses;
- CM1: Introduction of the random effect on the residual error reduced but not eliminated the bias;
- When subjects with the highest η_ε were incrementally removed from the datasets, bias due to non-compliance was further reduced and then eliminated (Figures 1 and 2). At the same time, η_ε variance decreased and then disappeared.
- For the datasets with all non-compliant subjects , CM1 method was not able to reduce the parameter bias. However, use of the pill count (fraction of missing doses) as a correction for the bioavailability of outpatient doses decreased or eliminated bias in clearance estimates;

Figure 1: Method CM1, sparse data, 50% non-compliant subjects, 60% missing doses, 2-day drug holidays

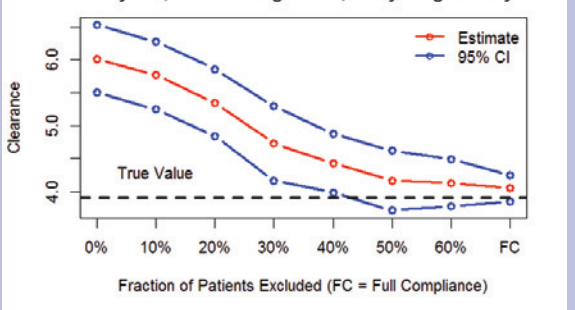
Estimate
95% CI
Clearance
True Value
4.0
5.0
6.0
0%
10%
20%
30%
40%
50%
60%
FC
Fraction of Patients Excluded (FC = Full Compliance)

Figure 2: Method CM1, sparse data, 50% non-compliant subjects, 60% missing doses, 2-day drug holidays

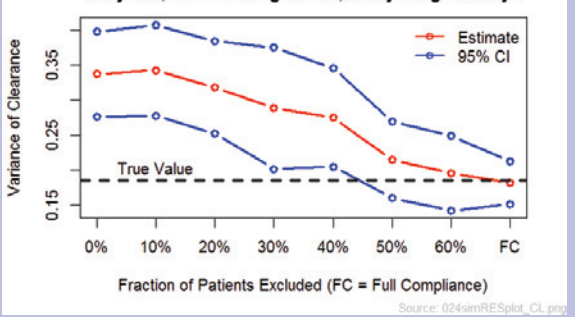
Estimate
95% CI
Variance of Clearance
True Value
0.15
0.25
0.35
0%
10%
20%
30%
40%
50%
60%
FC
Fraction of Patients Excluded (FC = Full Compliance)

- CM2 method provided unbiased estimates of the model parameters in the datasets with any fraction of non-compliant subjects. Simulated non-compliant subjects were estimated to have low bioavailability during outpatient dosing.

DISCUSSION AND CONCLUSIONS

For a number of simulated datasets with various sampling schemes and various fractions of non-compliant patients, the proposed methods allowed to identify subjects with compliance problem and to obtain the unbiased estimates of model parameters. These methods offer a way to evaluate the influence of compliance on the population PK parameter estimates.

CM1 and CM2 methods can be viewed as complementary, each with its own advantages and limitations. CM2 provides unbiased parameter estimates for any non-compliance pattern but it can be applied only for the specific sampling schemes that include relatively rich data following inpatient (fully compliant) doses. CM1 method is not based on any assumptions about the sampling schemes but it is unlikely to account for the non-compliance if it is present in the majority of patients. For the datasets with combined sparse and rich data, CM1 and CM2 method can be combined to provide the best results.

REFERENCES

[1] Karlsson MO, Jonsson EN, Wiltse CG, Wade JR, Assumption testing in population pharmacokinetic models: illustrated with an analysis of moxonidine data from congestive heart failure patients. J Pharmacokinet Biopharm. 1998 Apr; 26(2):207-46

[2] Gupta P, Hutmacher MM, Frame B, Miller R, An alternative method for population pharmacokinetic data analysis under noncompliance. J Pharmacokinet Pharmacodyn. 2008;35(2):219-33.