W5344

Immediate and Long-term Effects of Tocilizumab on Neutrophil Counts in Pediatric Patients With Systemic Juvenile Idiopathic Arthritis

Leonid Gibiansky,[1] Olivier Harari,[2] Nicolas Frey[3]

[1]QuantPharm LLC, [2]Roche Products Ltd, [3]F. Hoffmann-La Roche Ltd.

ABSTRACT

Purpose: Tocilizumab (TCZ) is a recombinant humanized IL-6 receptor monoclonal antibody that inhibits binding of IL-6 to its receptors. The aim of the analysis was to describe the time course of peripheral neutrophil counts (PNCs) after TCZ administration in patients with active systemic juvenile idiopathic arthritis (sJIA).

Methods: Serum TCZ concentrations and PNCs were available from 75 patients who received 12 mg/kg (patients <30 kg) or 8 mg/kg (patients ≥30 kg) infusions of TCZ or placebo every 2 weeks (total of 6 doses). Neutrophil counts were assessed at screening, baseline (week 0), and at 1, 2, 3, 6, 8, 10, and 12 weeks. A previously developed 2-compartment model with parallel linear and Michaelis-Menten elimination described TCZ concentrations. Different direct and indirect response models were tested to characterize the TCZ-PNC relationship.

Results: The TCZ-PNC relationship was described by a model that included an immediate TCZ effect on PNC decline (possibly, neutrophil margination) and a longer-term TCZ effect on PNC decline (toward normal levels) due to the improvement of patient condition (eg, decrease of inflammation). The immediate effect was described by a direct sigmoid E_{max} model (E_{max} = 0.724, EC_{50} = 6.38 µg/mL). These parameters were very similar to the respective values obtained earlier for adult patients (E_{max} = 0.788, EC_{50} = 7.49 µg/mL). The maximum rate of decline of the long-term effect was 0.166 day^{-1} and the TCZ concentration corresponding to half of this rate was 151 µg/mL. The corresponding PNC decline for a typical patient was estimated to go from 8.12 × 10^9/L to 5.72 × 10^9/L. The magnitude of the decline increased with the baseline concentrations of the C-reactive protein. Diagnostic plots and predictive check simulations indicated a good agreement of model predictions with the observed data.

Conclusions: The PNC time course following Q2W administration of 12 mg/kg (for patients <30 kg) or 8 mg/kg (for patients ≥30 kg) of TCZ in pediatric patients with sJIA was characterized by a combination of an immediate response described by the sigmoid E_{max} function of TCZ concentrations and a slow decline possibly related to the improvement of the patient condition.

BACKGROUND

- Tocilizumab (TCZ) is a recombinant humanized interleukin-6 (IL-6) receptor monoclonal antibody that inhibits binding of IL-6 to its receptors

PURPOSE

- The aim of the analysis was to describe the time course of peripheral neutrophil counts (PNCs) after TCZ administration in a pediatric population with active systemic juvenile idiopathic arthritis (sJIA)

METHODS

- Serum TCZ concentrations and PNCs were available in 75 pediatric patients with active sJIA who received 12 mg/kg (for patients <30 kg) or 8 mg/kg (for patients ≥30 kg) infusions of TCZ every 2 weeks (total of 6 doses)
- PNCs were assessed at screening, at baseline (week 0), and at 1, 2, 3, 6, 8, 10, and 12 weeks
- A previously developed 2-compartment model with parallel linear and Michaelis-Menten elimination described TCZ concentrations[1,2]
- Different pharmacokinetic (PK)/pharmacodynamic (PD) models with direct and indirect responses were tested to characterize the TCZ–PNC relationship

RESULTS

- The TCZ–PNC relationship was described by a model that included an immediate TCZ effect on PNC decline (possible increase of neutrophil margination)[3] and a longer-term TCZ effect on PNC decline (toward normal levels) due to improvement in patient condition (eg, decrease in inflammation)
- The immediate effect was described by a direct sigmoid E_{max} model (E_{max} = 0.724 [% relative standard error (RSE) 14.8%] and a half-maximal effective concentration [EC_{50}] = 6.38 µg/mL [%RSE 15.8%])
- The PK/PD parameters were very similar to the respective values obtained earlier for adult patients[2] (E_{max} = 0.788 and EC_{50} = 7.49 µg/mL)
- The maximum rate of decline of the long-term effect was 0.166 day^{-1}, and the TCZ concentration inducing half this rate was 151 µg/mL
- The corresponding PNC decline for a typical patient was estimated to go from 8.12 × 10^9/L to 5.72 × 10^9/L
- The magnitude of the decline increased with the baseline concentration of C-reactive protein (CRP)
- Diagnostic plots and predictive check simulations indicated good agreement of model predictions with observed data

Figure 1. Observed PNC data versus nominal time.

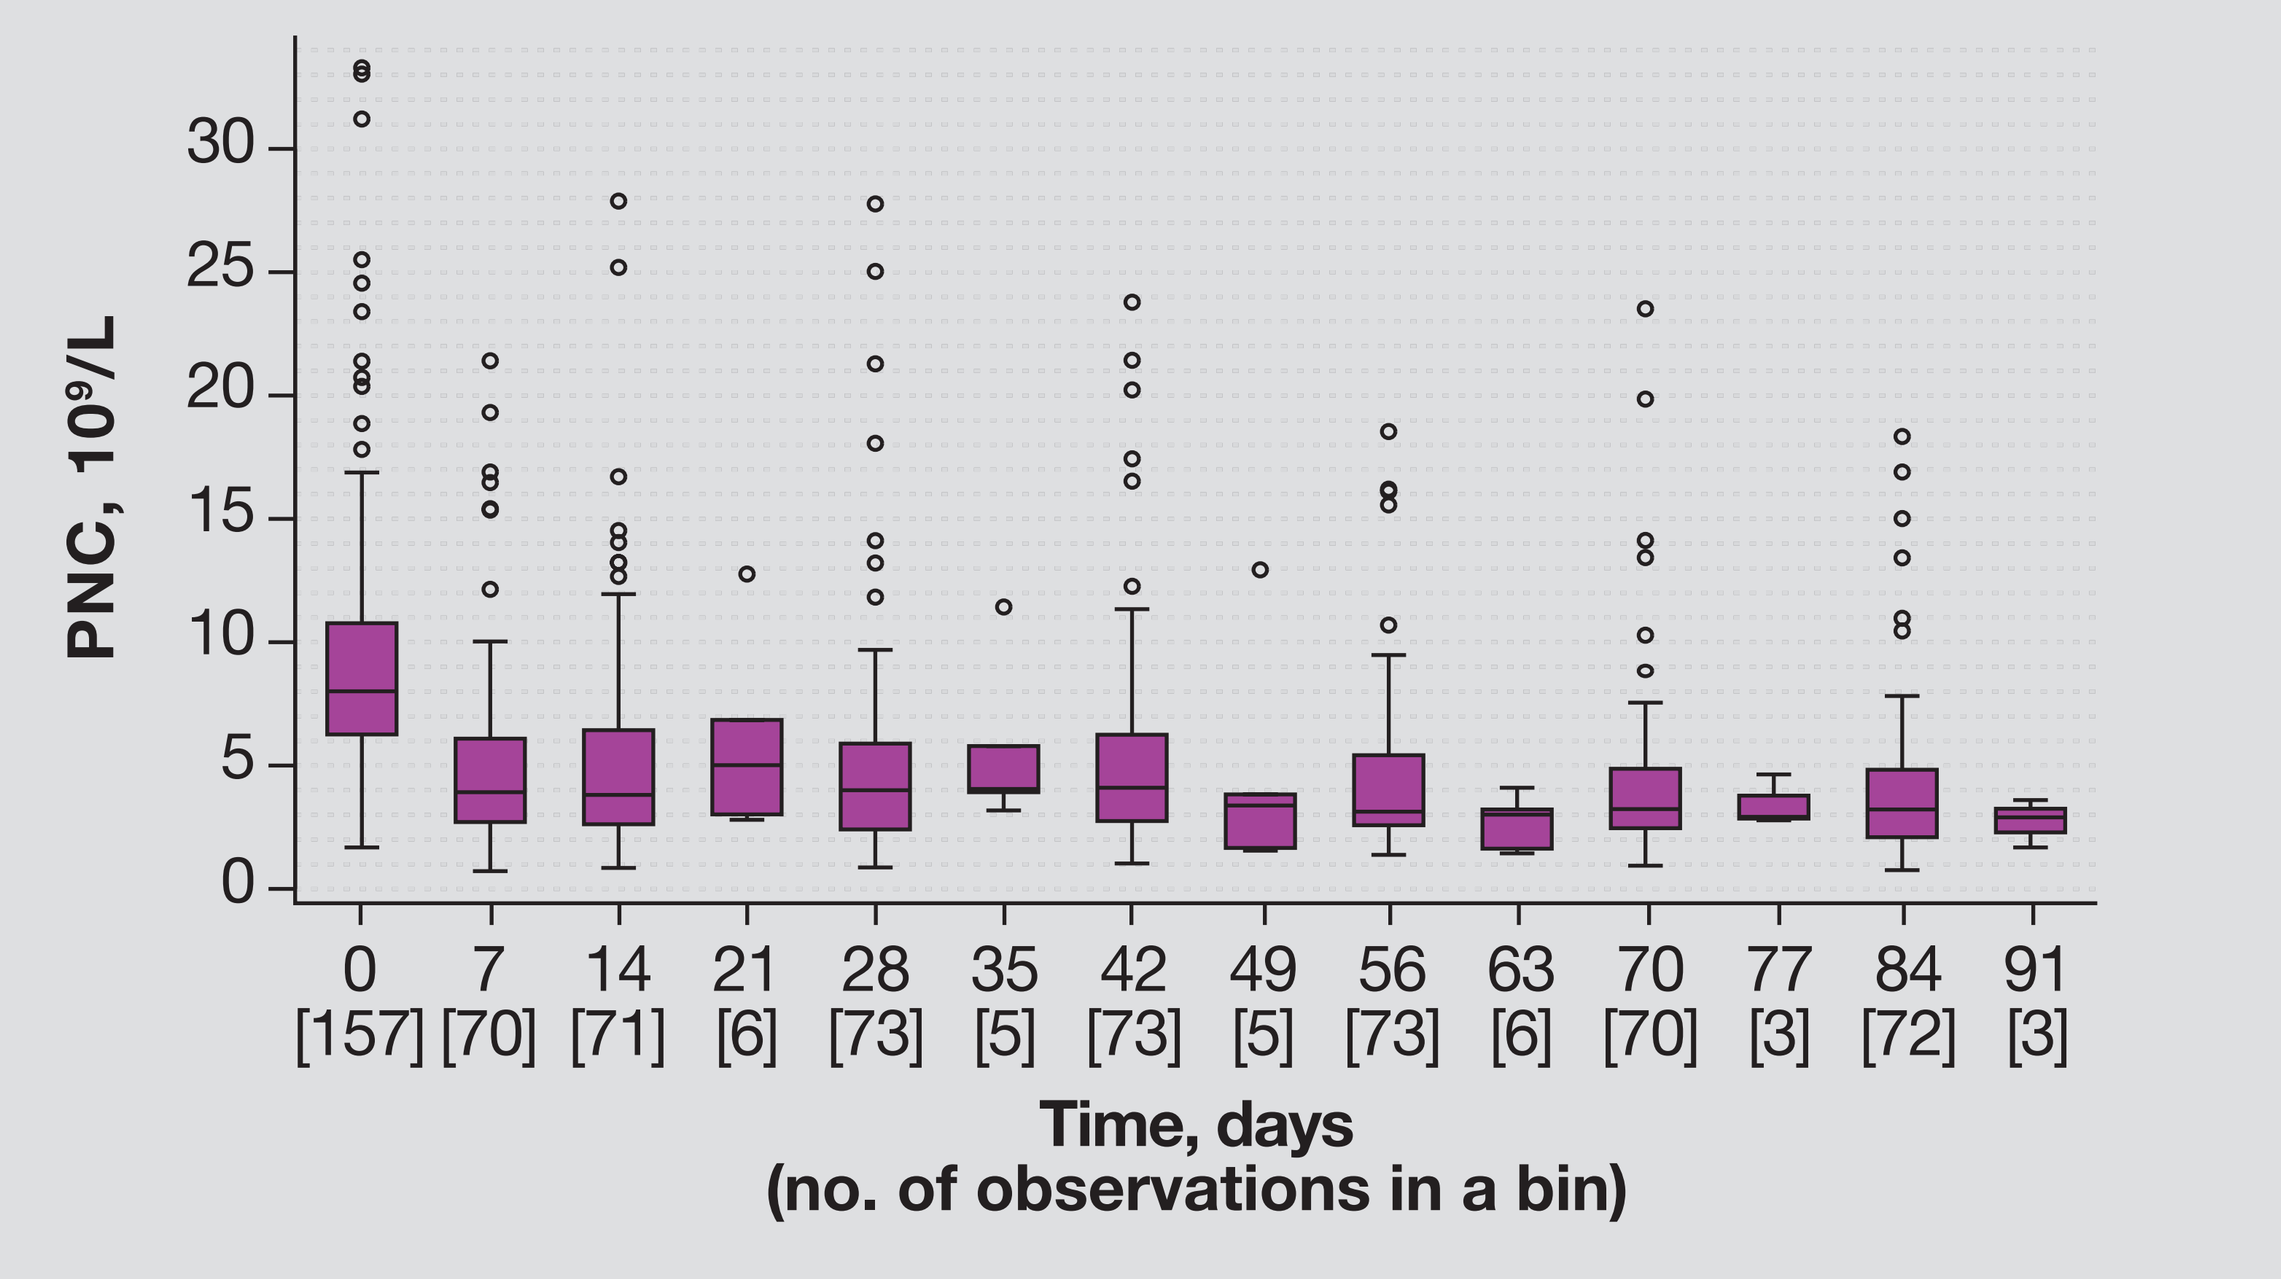

PNC, 10⁹/L
0 7 14 21 28 35 42 49 56 63 70 77 84 91
[157] [70] [71] [6] [73] [5] [73] [5] [73] [6] [70] [3] [72] [3]
Time, days
(no. of observations in a bin)

TCZ Concentration—PNC Model

$$PNC_{total} = PNC_1 + PNC_2$$

$$\frac{dPNC_2}{dt} = K_{out}(BPNC_2 - PNC_2) - K(C) \times PNC_2 \qquad K(C) = \frac{K_{max} \times C^{\gamma 1}}{K_{50}^{\gamma} + C^{\gamma 1}}$$

$$PNC_{circ} = \frac{PNC_{total}}{1 + H(C)} \qquad H(C) = \frac{E_{max} \times C^{\gamma}}{EC_{50}^{\gamma} + C^{\gamma}} \qquad BPNC_2 = \theta_2 \left(\frac{BCRP}{100}\right)^{BPNC_2,\ CRP}$$

Figure 2. Schematic representation of the final TCZ-PNC model. The pink shaded region shows the observed quantity. The model is described in the text. K_{out} was small and poorly estimated; it was fixed to zero in the final model.

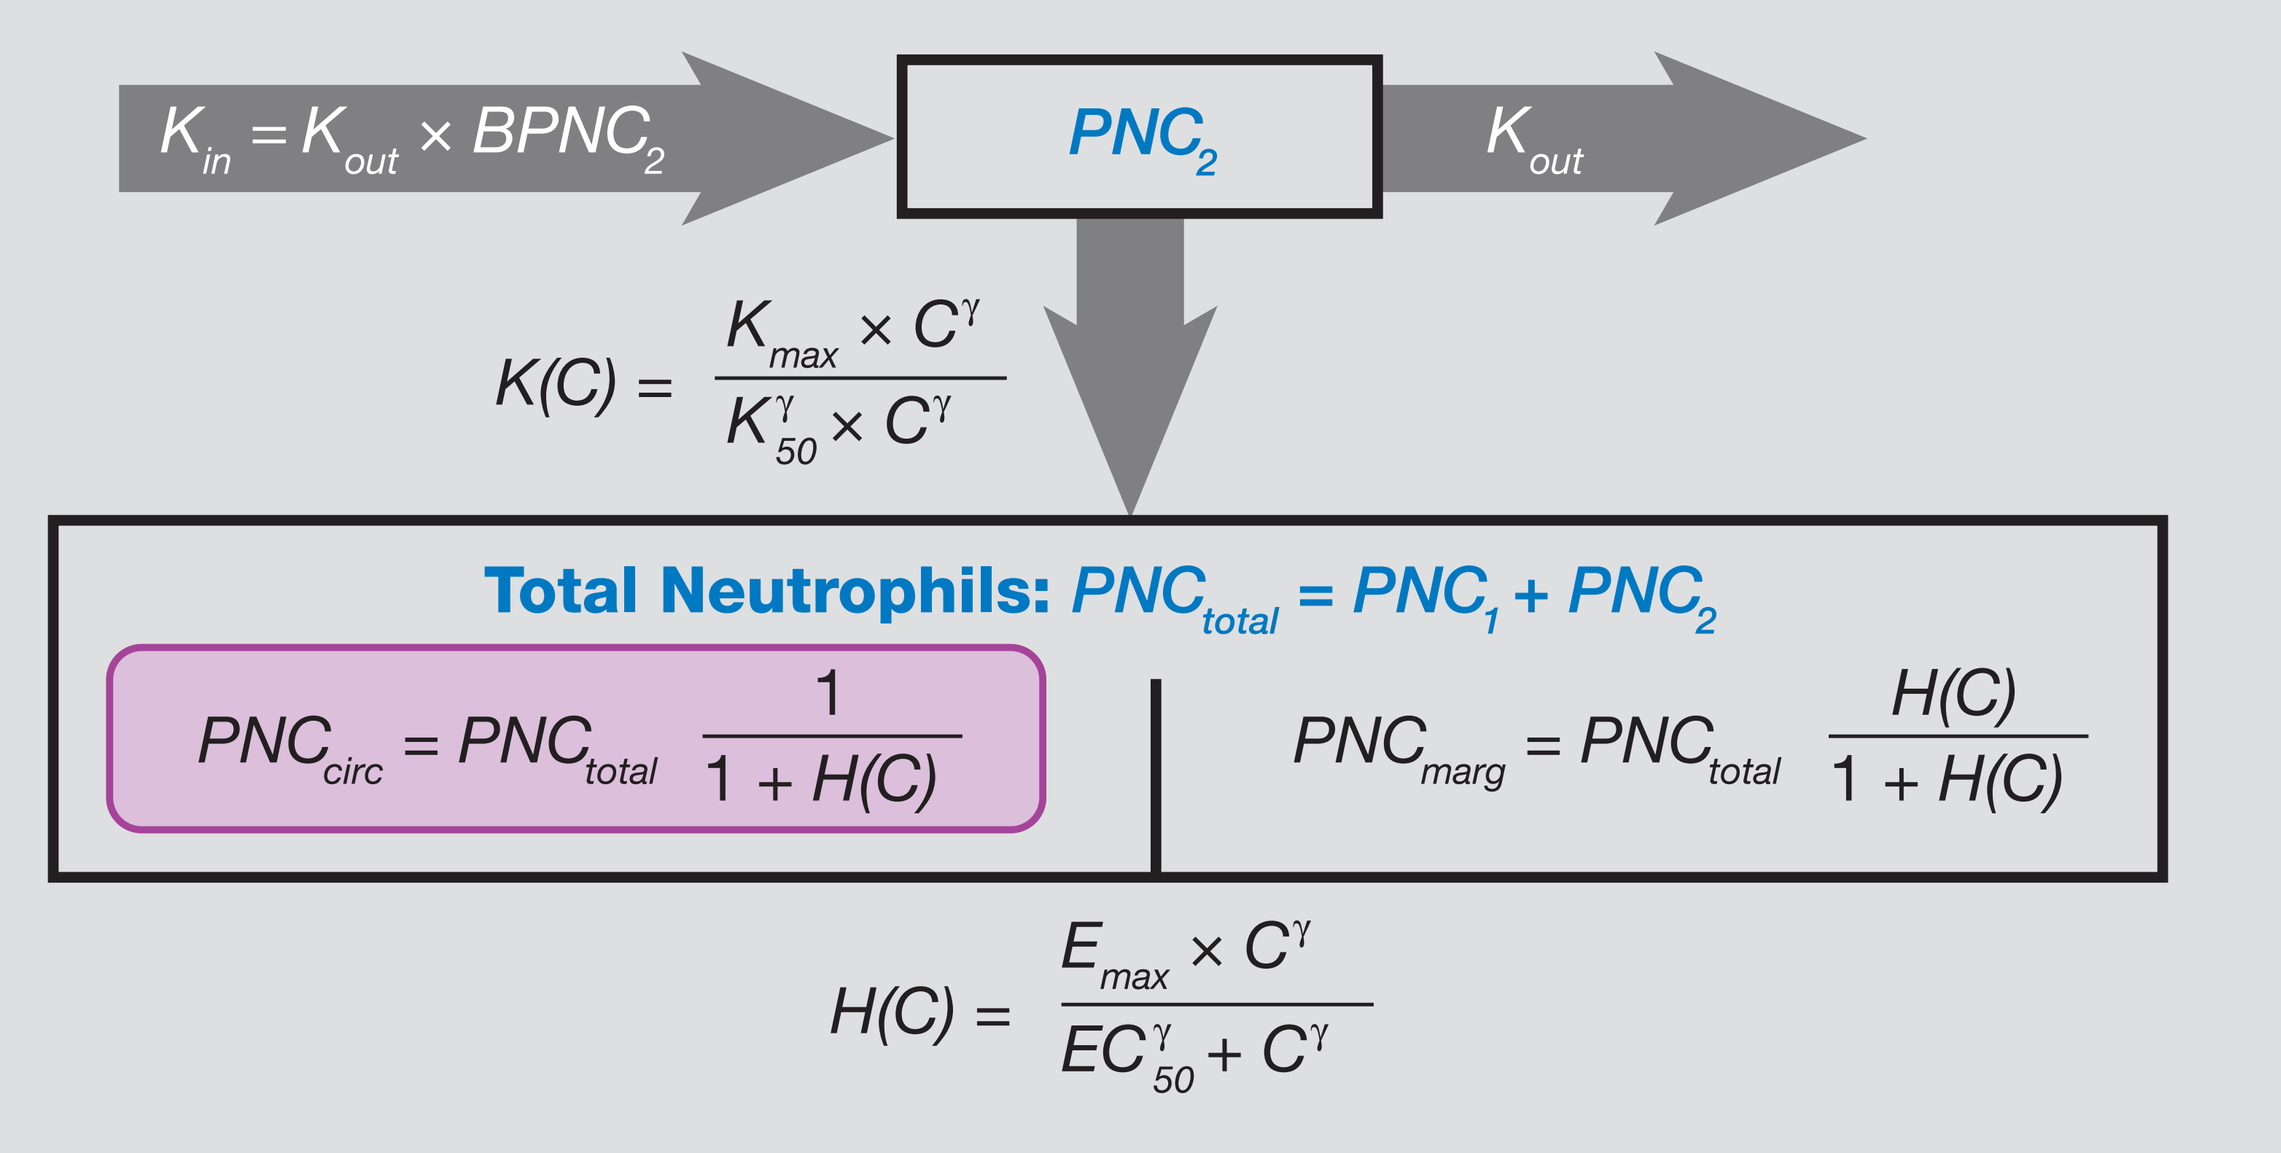

K_in = K_out × BPNC_2
PNC_2
K_out
K(C) = K_max × C^γ / K^γ_50 × C^γ
Total Neutrophils: PNC_total = PNC_1 + PNC_2
PNC_circ = PNC_total 1 / 1 + H(C)
PNC_marg = PNC_total H(C) / 1 + H(C)
H(C) = E_max × C^γ / EC^γ_50 + C^γ

- PNC_{total} is the sum of observed circulating neutrophils (PNC_{circ}) and unobserved marginalized neutrophils (PNC_{marg}). PNC_{total} slowly decreases with time on TCZ due to the decrease of PNC_2 from its baseline level, $BPNC_2$. $BPNC_2$ was higher in patients with high CRP (high inflammation)
- K_{out} was estimated to be small and was fixed to zero

Table 1. Parameter Estimates for the Final Model

Parameter		Estimate (95% CI)	%RSE	Bootstrap Median (95% CI)	Variability	Shrinkage
PNC_1 (10^9/L)	θ_1	5.72	13.4	4.22, 7.21		
$BPNC_2$ (10^9/L)	θ_2	2.40	30.2	0.981, 3.82		
K_{50} (µg/mL)	θ_3	151	66.1	0, 347		
K_{max}	θ_4	0.166	244	–0.627, 0.959		
EC_{50} (µg/mL)	θ_5	6.38	15.8	4.4, 8.36		
E_{max}	θ_6	0.724	14.8	0.514, 0.935		
γ (≤5)	θ_7	5		At upper bound		
$RATIO_{ETA}$	θ_8	0.579	35.1	0.18, 0.977		
$BPNC_{2,CRP}$	θ_9	0.523	59.8	–0.09, 1.14		
ω^2_{PNC1}	Ω (1, 1)	0.315	31.0	0.124, 0.506	CV = 56.1%	9.8%
R ω_{PNC1} ω_{PNC2}	Ω (2, 1)	–0.391	59.5	–0.846, 0.0646	R = –0.638	
ω^2_{PNC2}	Ω (2, 2)	1.19	45.5	0.129, 2.25	CV = 109%	28.0%
ω^2_{Emax}	Ω (3, 3)	2.10	70.5	0, 5	CV = 145%	17.7%
σ_{prop}^2	Σ (1, 1)	0.0961	5.22	0.0863, 0.106	CV = 31.0%	9.1%

95% CI, 95% confidence interval; CV, coefficient of variation (CV = 100 × SD%); PE, parameter estimate; RSE, relative standard error (RSE = 100 × SE/PE); SD, standard deviation; SE, standard error.

Model Validation (VPC) and Model-based Simulations

Figure 3. Visual predictive check for model 310: PNC versus time, all patients and by nominal TCZ dose. The lines show median (purple) and the 10th and 90th percentiles (blue) of the observed PNCs. The shaded regions show the 80% CIs on these quantities obtained by simulations. The simulated values were computed from 500 trials simulated using dosing, sampling, and the covariate values of the analysis data set.

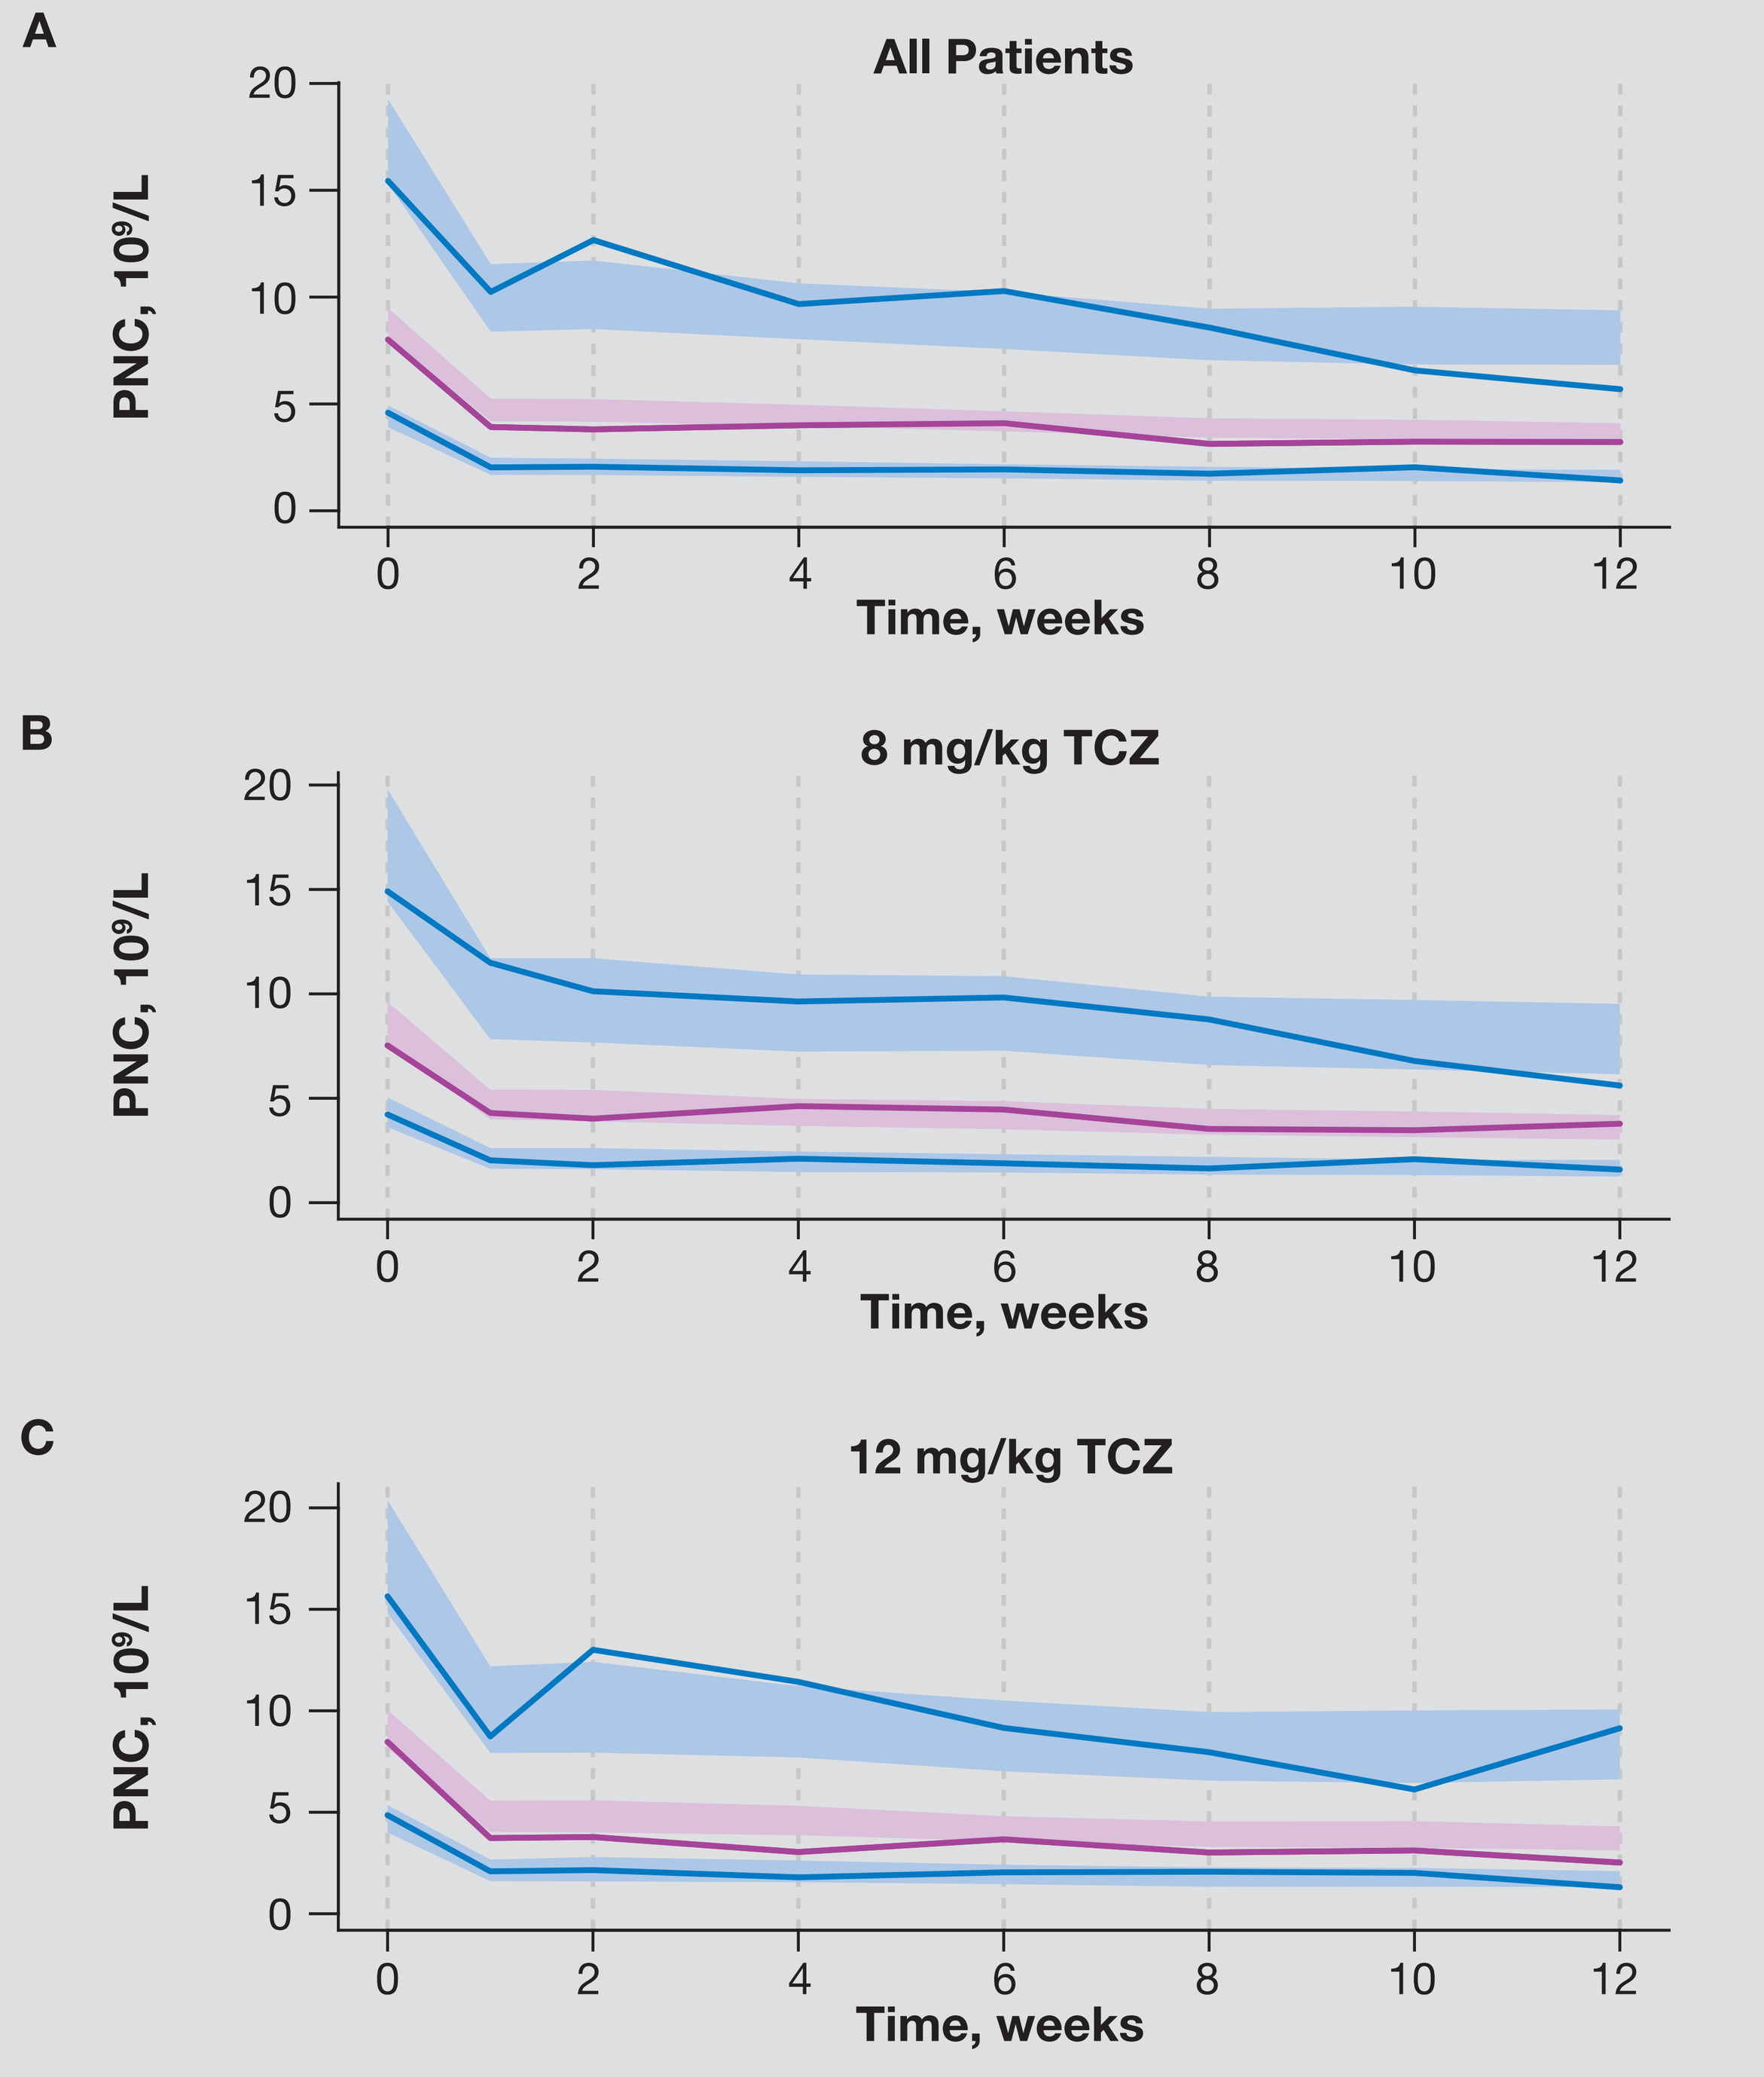

A
All Patients
B
8 mg/kg TCZ
C
12 mg/kg TCZ
PNC, 10⁹/L
Time, weeks

Figure 4. Model-based simulation results by TCZ dose (weight [WT]) group. (A) Blue lines correspond to 90% coverage intervals of the simulated PNCs. Purple lines show medians of the simulated values. Gray lines show the normal range for PNCs (1.5 to 8 × 10^9/L). (B) Blue lines correspond to 90% coverage intervals of the individual predictions of the TCZ concentration in the analysis population. Purple lines show medians of these values.

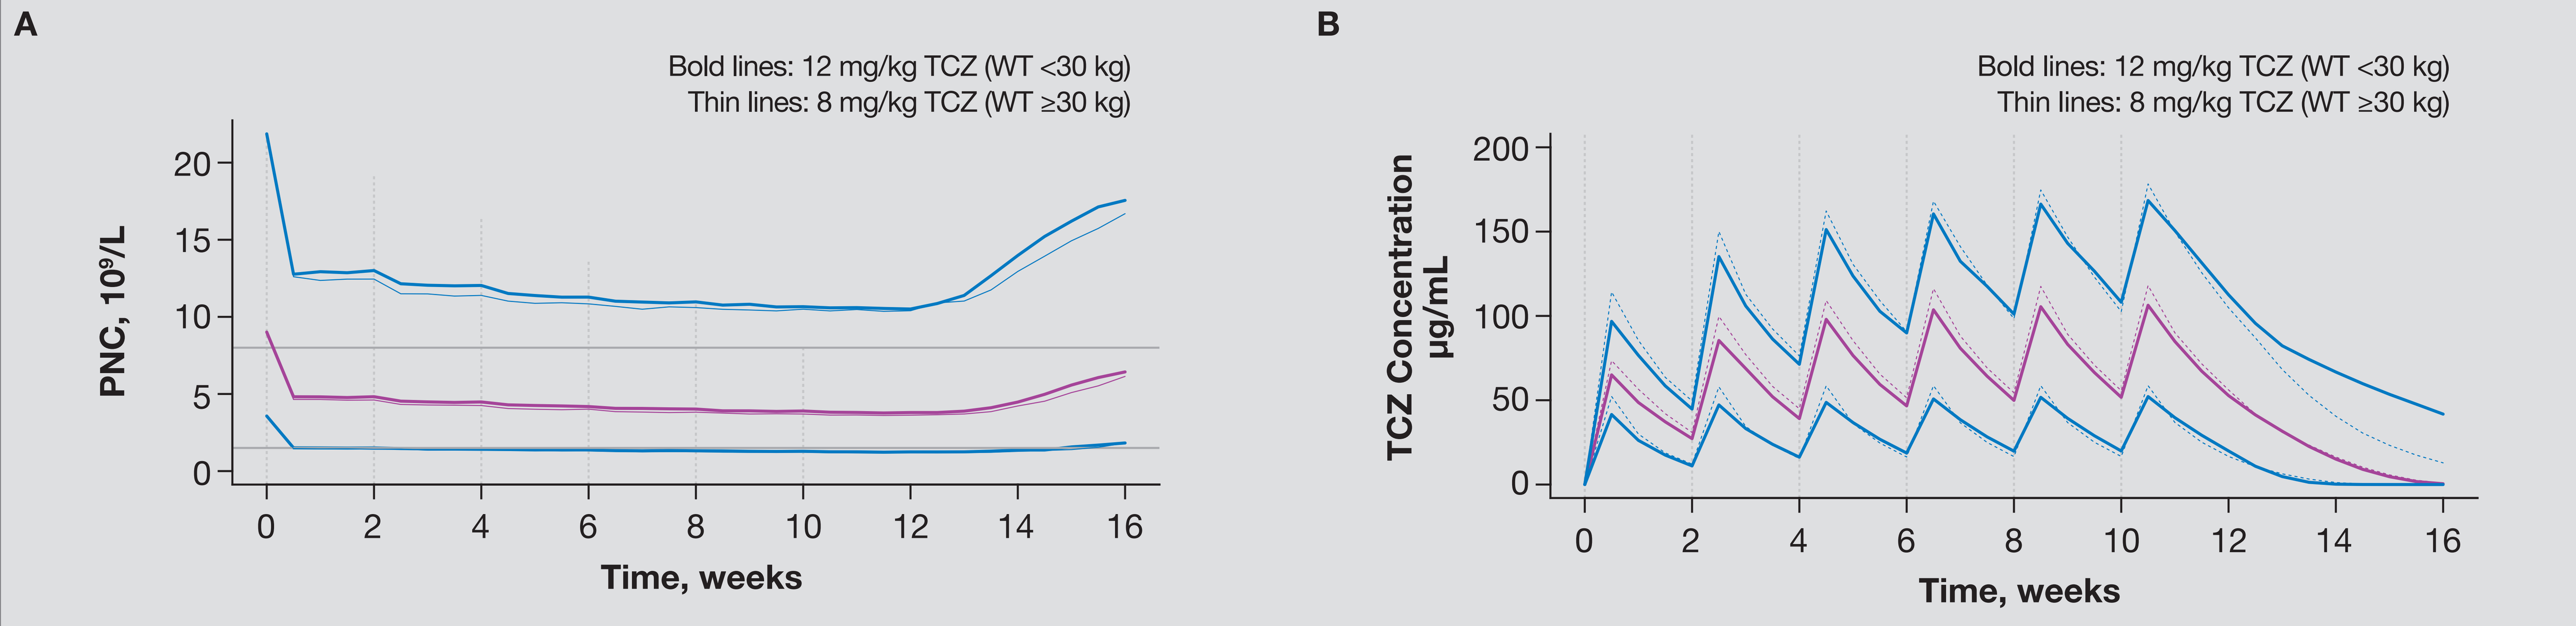

A
B
Bold lines: 12 mg/kg TCZ (WT <30 kg)
Thin lines: 8 mg/kg TCZ (WT ≥30 kg)
PNC, 10⁹/L
TCZ Concentration µg/mL
Time, weeks

Figure 5. Basic goodness-of-fit plots for the final model.

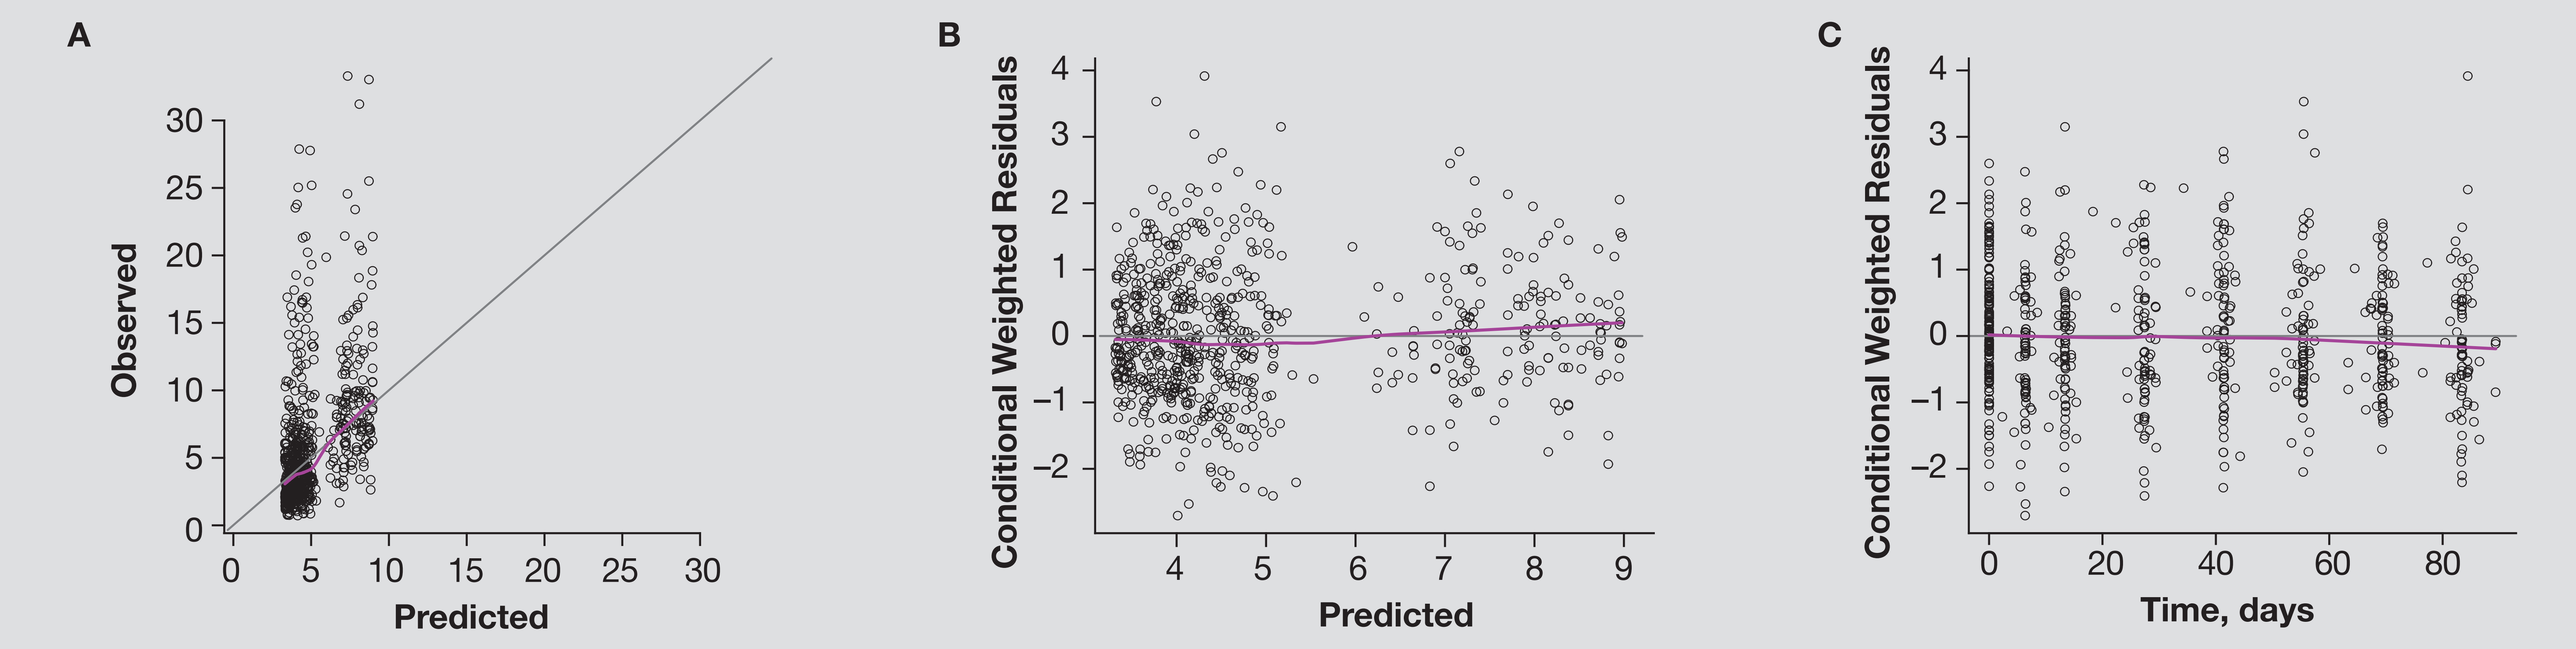

A
B
C
Observed
Predicted
Conditional Weighted Residuals
Time, days

Figure 6. Model-based simulation results: PNC_2 time course.

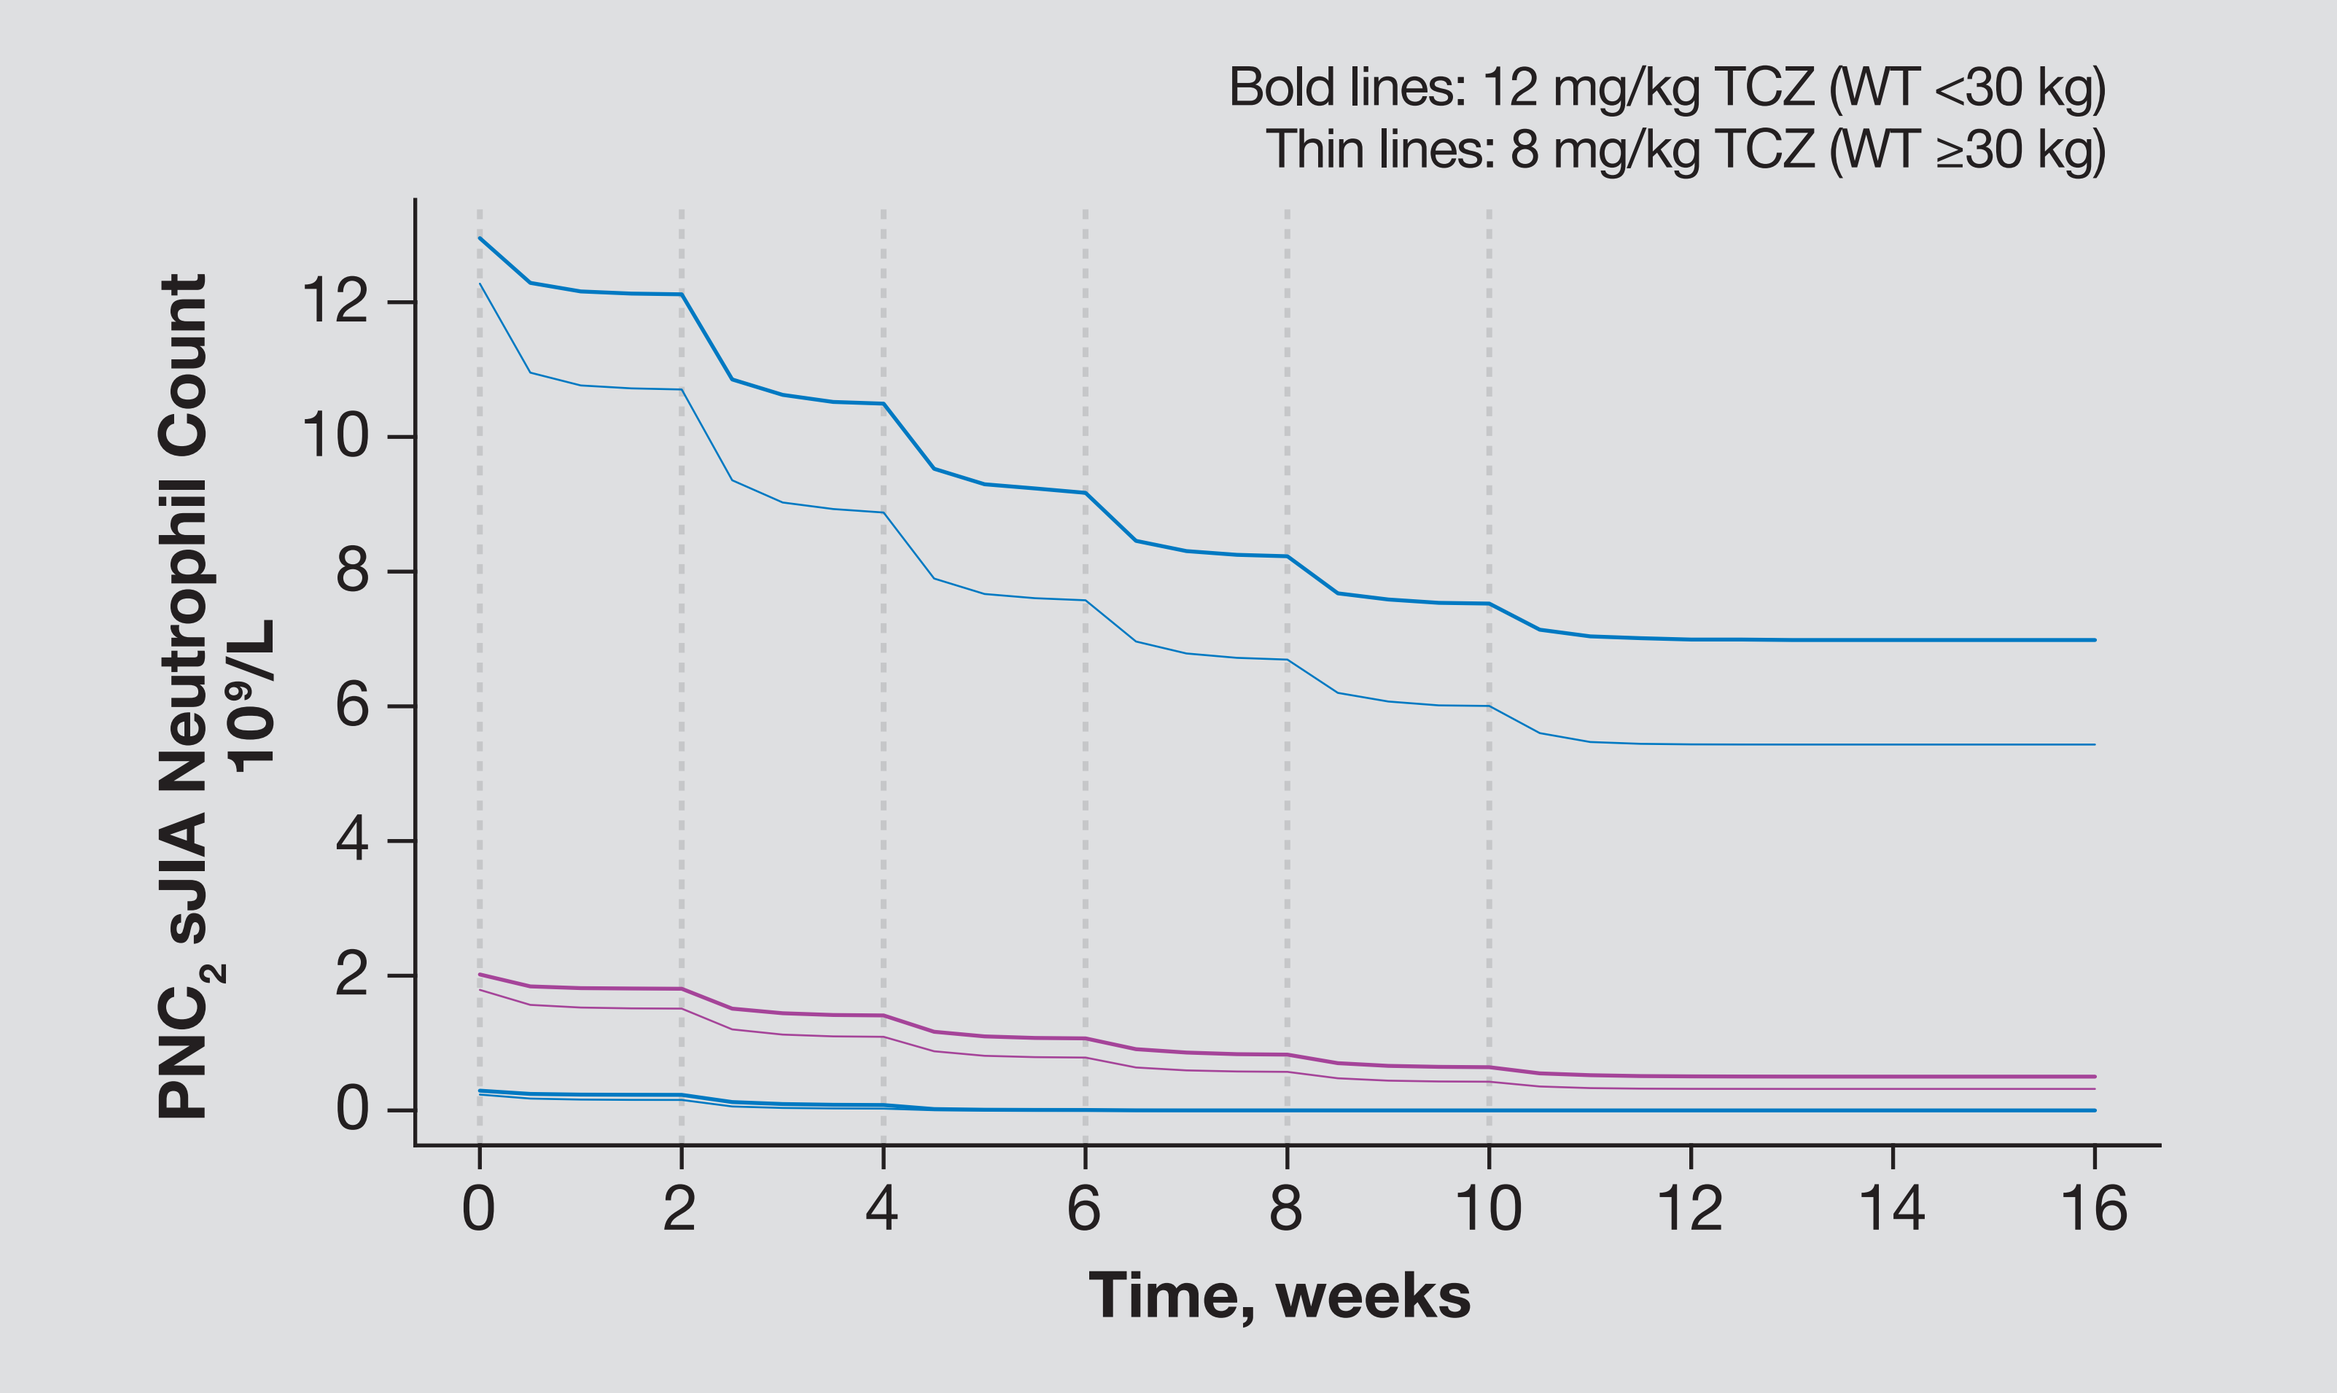

Bold lines: 12 mg/kg TCZ (WT <30 kg)
Thin lines: 8 mg/kg TCZ (WT ≥30 kg)
PNC2 sJIA Neutrophil Count 10⁹/L
Time, weeks

CONCLUSION

- The observed changes in neutrophil data are consistent with the TCZ mechanism of action and can be fully explained by a short-term effect assuming neutrophil margination and a long-term effect assuming improvement in patient condition (eg, decrease in inflammation)

REFERENCES

1. Frey N et al. *J Clin Pharmacol.* 2010;50:754-766.
2. Roche internal documents.
3. Gibiansky L, Frey N. *J Pharmacokinet Pharmacodyn.* 2012;39:5-16.

To receive a PDF of this poster on your mobile phone:
- Go to getscanlife.com from your mobile browser to download the free barcode reader application
- Scan the code and get access to content

http://bit.ly/QFEPwU (This URL is case sensitive)

Presented at the 2012 American Association of Pharmaceutical Scientists Annual Meeting; October 14–18, 2012; Chicago, Illinois

This study was funded by Roche.
Support for third-party editorial assistance for this poster was provided by F. Hoffmann-La Roche Ltd.